Title: Expanding Tissue Donor Referrals: Implementation of an EMS Referral Program for Non-Hospital Deaths

Authors: Alexander Pajkowski, Medical Examiner & Funeral Home Liaison; Stephanie Sommer, Manager of Partner Relations; Mary Pat Butcher, Director of Hospital Development

Background

The state requires medical examiners to report non-hospital deaths to the organ procurement organization (OPO) within a reasonable timeframe. In larger, well-funded counties, electronic case management systems integrate with the OPO's systems, enabling automatic referrals upon entry by death investigators. However, in smaller, rural counties with limited resources, a single physician often assumes full responsibility for death investigations and communications, frequently without dedicated support staff. These counties face minimal regulatory pressure to collaborate with the OPO, as current laws offer little enforcement or incentive. As a result, many individuals who die outside of hospitals are never referred, and their families are not offered the opportunity for tissue donation discussions. In contrast, all hospitals are federally required by CMS regulations to report deaths within one hour, a standard that is consistently met. This disparity created a critical need to engage alternative referral sources for non-hospital deaths.

Hypothesis

Rather than relying solely on medical examiners to report non-hospital deaths, we hypothesized that emergency medical services (EMS) agencies could serve as an alternative source of referral information. Because EMS personnel are routinely dispatched to scenes involving non-hospital deaths, we anticipated that access to EMS reports would allow the OPO to identify potential tissue donors in real-time. Our goal was to establish a method for systematically receiving and reviewing EMS reports to supplement or replace traditional medical examiner referrals where gaps existed.

Methods

We partnered with the State Division of EMS and identified that all EMS reports statewide

are aggregated into a centralized database. Collaborating with the database development team, we implemented a custom filter to extract EMS reports from selected counties specifically marked as a 'death on scene.' To securely receive these reports, we established a Secure File Transfer Protocol (SFTP) system, allowing

filtered EMS reports to be transmitted in their raw XML format, structured according to the National Emergency Medical Services Information System (NEMSIS) standard. An automated internal process was developed to parse the incoming XML data, translate it into a readable format, and compile the essential clinical and demographic information. This information is then delivered via automated email to OPO staff, allowing it to serve as a substitute referral pathway for non-hospital deaths.

Results

Between January 2024 and January 2025, a total of 641 unique non-hospital deaths were identified through the EMS referral pathway. Reports were successfully attributed across multiple rural counties, with the most populous county accounting for 116 cases, followed by the next most populous counties reporting 94 and 72 cases respectively. However, only approximately 150 referrals were received within an actionable timeframe, due in part to delays in EMS personnel report writing and a 12-hour transmission lag from a widely used intermediary database system. Despite these challenges, 17 individuals were able to become tissue donors as a direct result of the EMS referral program. This alternative referral strategy increased access to donation opportunities for families who might otherwise not have been contacted under traditional medical examiner-dependent models.

Conclusions

The implementation of an EMS-based referral pathway demonstrated that non-hospital deaths could be identified outside of traditional medical examiner channels, resulting in increased tissue donation opportunities. Although systemic delays in report transmission limited the

number of timely referrals, the program successfully led to 17 additional tissue donors during the pilot period. These findings suggest that further collaboration with EMS systems, targeted education for EMS personnel regarding timely report submission, and technical improvements to data transmission could significantly enhance the impact of this referral model. Broader adoption of EMS referrals across rural and under-resourced regions may offer a scalable solution to improve donor identification and maximize tissue donation nationwide.

Expanding Tissue Donor Referrals:

Implementation of an EMS Referral Program for Non-Hospital Deaths

Authors:
Alexander Pajkowski • Medical Examiner & Funeral Home Liaison
Stephanie Sommer • Manager of Partner Relations
Mary Pat Butcher • Director of Hospital Development

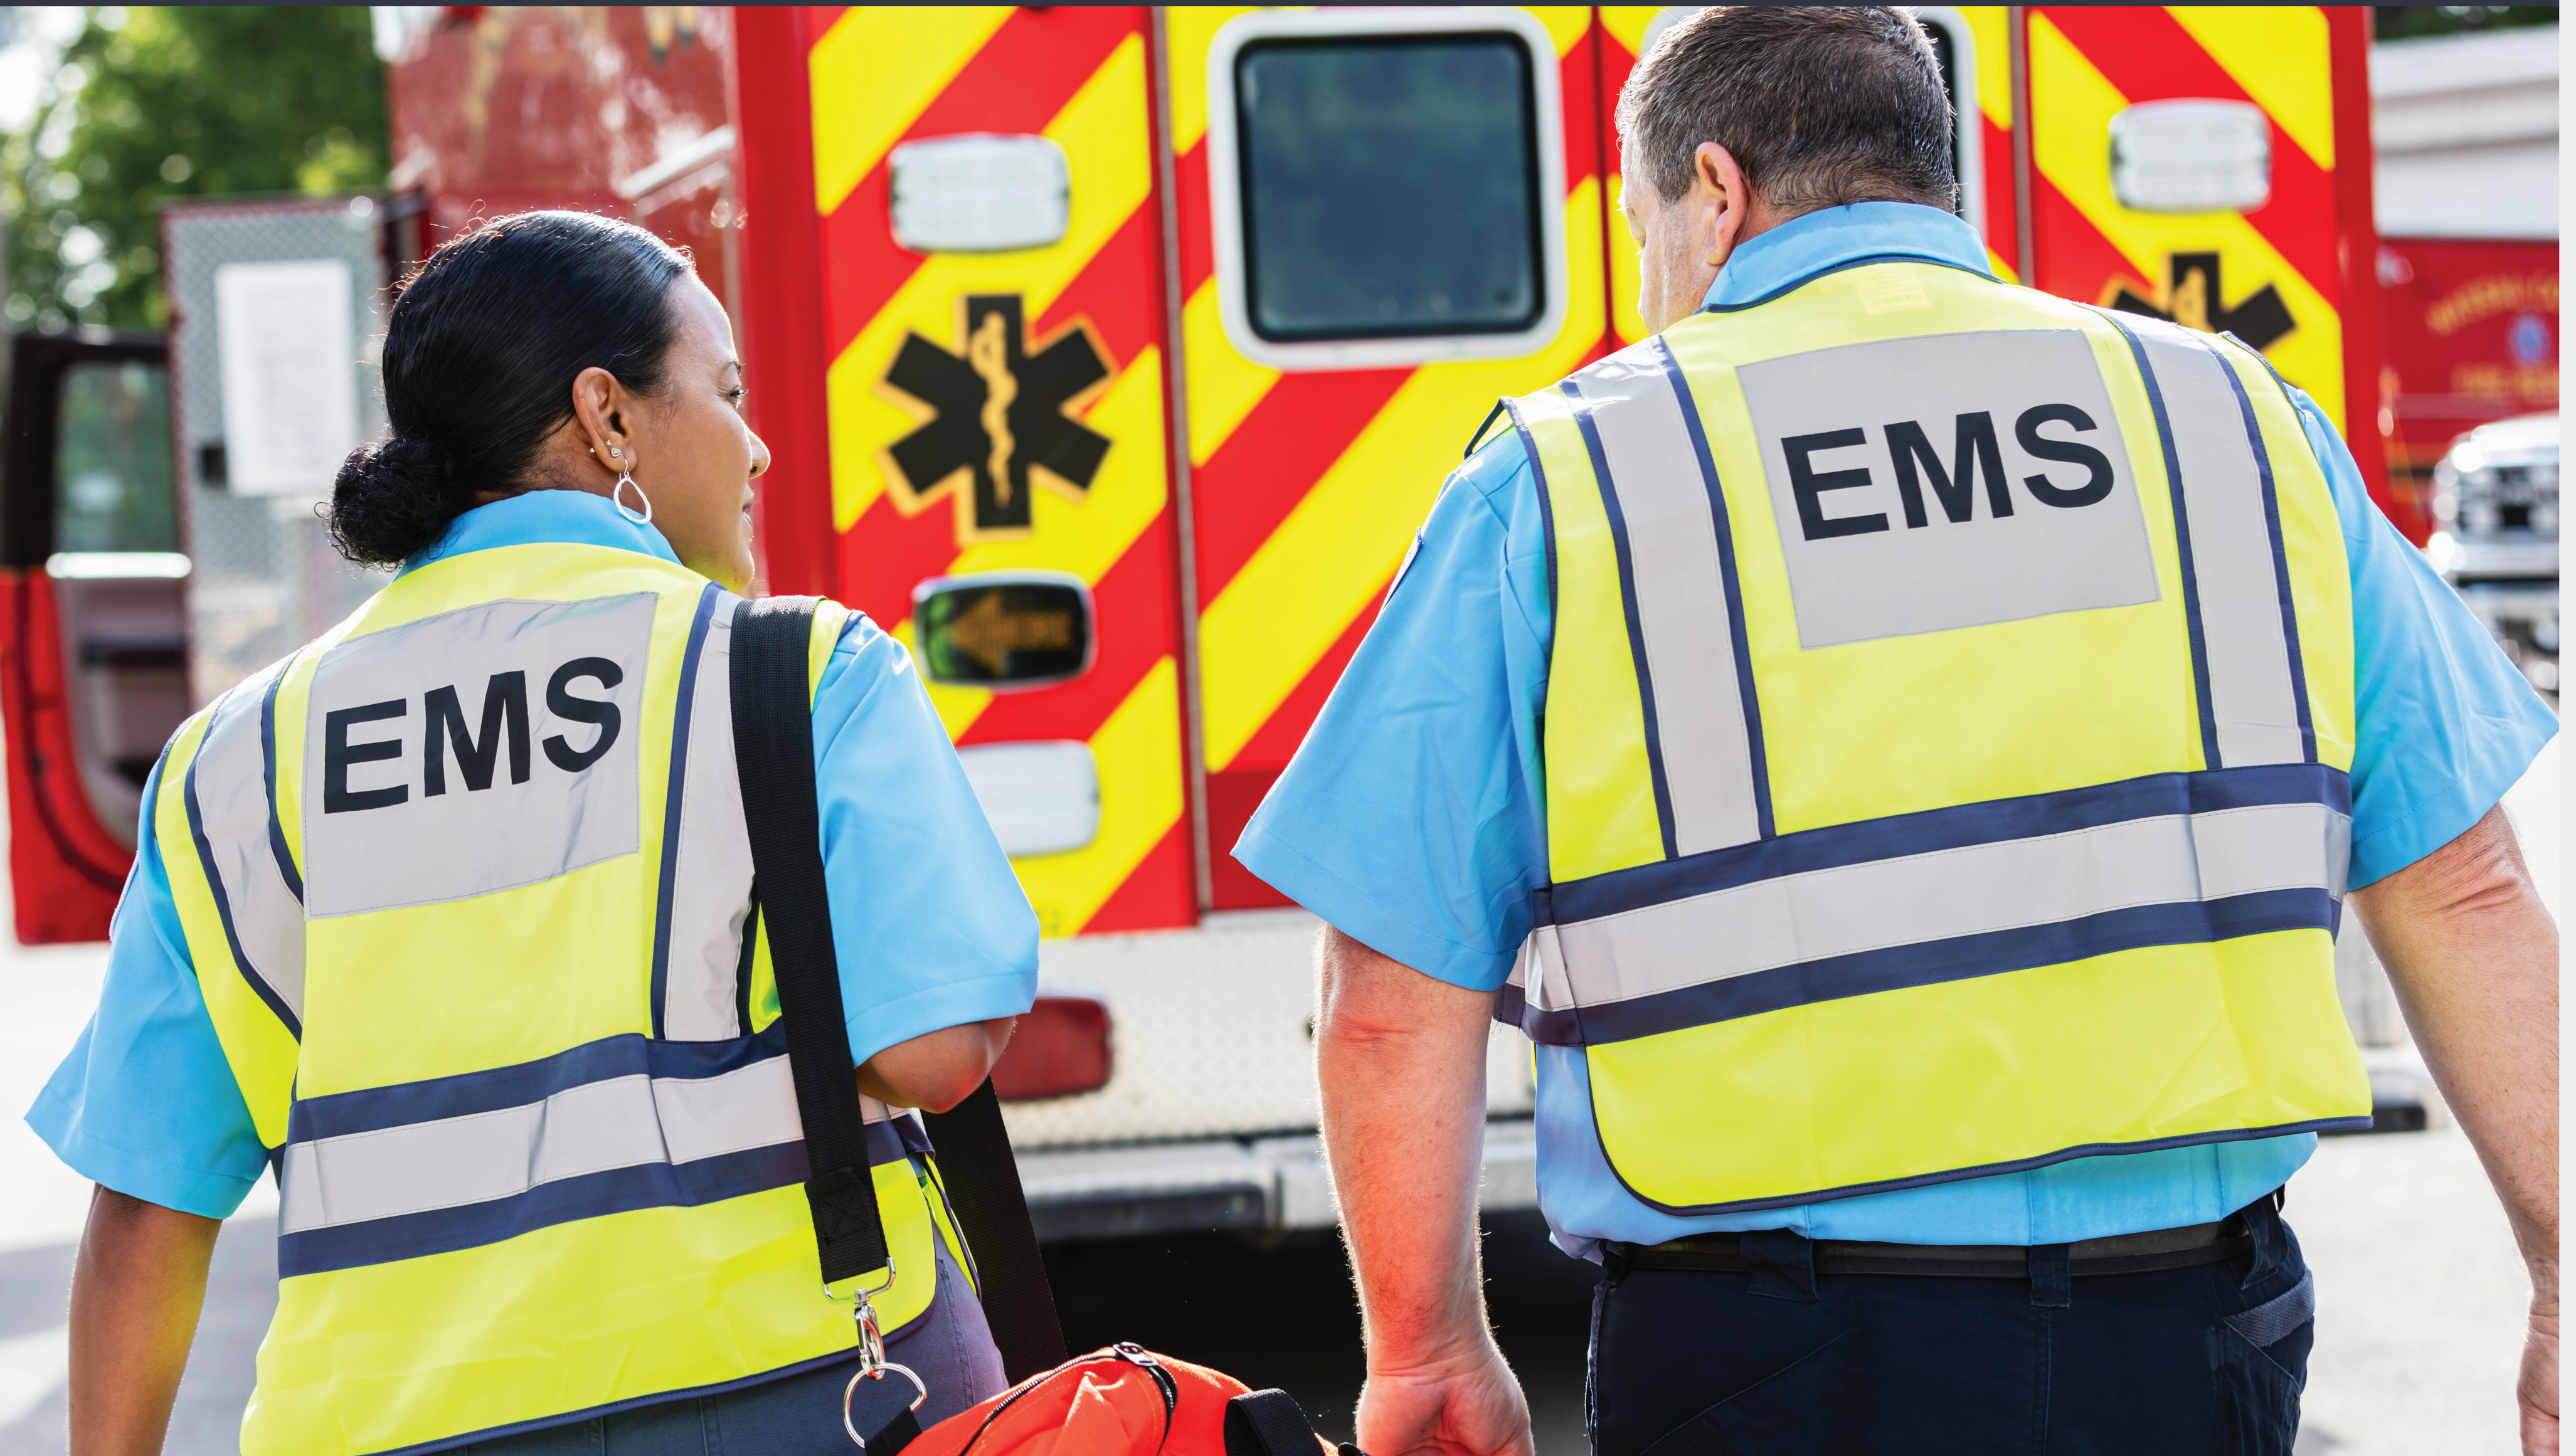
EMS
EMS

BACKGROUND

The state requires medical examiners to report non-hospital deaths to the organ procurement organization (OPO) within a reasonable timeframe. In larger, well-funded counties, electronic case management systems integrate with the OPO's systems, enabling automatic referrals upon entry by death investigators. However, in smaller, rural counties with limited resources, a single physician often assumes full responsibility for death investigations and communications, frequently without dedicated support staff. These counties face minimal regulatory pressure to collaborate with the OPO, as current laws offer little enforcement or incentive.

As a result, many individuals who die outside of hospitals are never referred, and their families are not offered the opportunity for tissue donation discussions. In contrast, all hospitals are federally required by CMS regulations to report deaths within one hour, a standard that is consistently met. This disparity created a critical need to engage alternative referral sources for non-hospital deaths.

HYPOTHESIS

Rather than relying solely on medical examiners to report non-hospital deaths, we hypothesized that emergency medical services (EMS) agencies could serve as an alternative source of referral information. Because EMS personnel are routinely dispatched to scenes involving non-hospital deaths, we anticipated that access to EMS reports would allow the OPO to identify potential tissue donors in real-time. Our goal was to establish a method for systematically receiving and reviewing EMS reports to supplement or replace traditional medical examiner referrals where gaps existed.

METHODS

We partnered with the State Division of EMS and identified that all EMS reports statewide are aggregated into a centralized database. Collaborating with the database development team, we implemented a custom filter to extract EMS reports from selected counties specifically marked as a 'death on scene.' To securely receive these reports, we established a Secure File Transfer Protocol (SFTP) system, allowing filtered EMS reports to be transmitted in their raw XML format, structured according to the National Emergency Medical Services Information System (NEMSIS) standard.

An automated internal process was developed to parse the incoming XML data, translate it into a readable format, and compile the essential clinical and demographic information. This information is then delivered via automated email to OPO staff, allowing it to serve as a substitute referral pathway for non-hospital deaths.

RESULTS

Between January 2024 and January 2025, a total of 641 unique non-hospital deaths were identified through the EMS referral pathway. Reports were successfully attributed across multiple rural counties, with the most populous county accounting for 116 cases, followed by the next most populous counties reporting 94 and 72 cases respectively. **However, only approximately 150 referrals were received within an actionable timeframe**, due in part to delays in EMS personnel report writing and a 12-hour transmission lag from a widely used intermediary database system. Despite these challenges, **17 individuals were able to become tissue donors as a direct result of the EMS referral program.**

This alternative referral strategy increased access to donation opportunities for families who might otherwise not have been contacted under traditional medical examiner-dependent models.

641
unique non-hospital deaths were identified

150
referrals were received within an actionable timeframe

17
individuals were able to become tissue donors

CONCLUSION

The implementation of an EMS-based referral pathway demonstrated that non-hospital deaths could be identified outside of traditional medical examiner channels, resulting in increased tissue donation opportunities. Although systemic delays in report transmission limited the number of timely referrals, the program successfully led to 17 additional tissue donors during the pilot period.

These findings suggest that further collaboration with EMS systems, targeted education for EMS personnel regarding timely report submission, and technical improvements to data transmission could significantly enhance the impact of this referral model. Broader adoption of EMS referrals across rural and under-resourced regions may offer a scalable solution to improve donor identification and maximize tissue donation nationwide.

GIFT of LIFE MICHIGAN SM
We honor life through donation.SM
golm.org